Early Response to Certolizumab Pegol in Rheumatoid Arthritis Predicts Outcome: Data from a Prospective Observational Study

A. Saraux,[1] R. M. Flipo,[2] F. Fagnani,[3] J. Massol,[4] B. Combe,[5] P. Huot-Marchand,[6] J-M. Joubert,[7] J. Dunkel,[8] G. Cukierman[7]

[1]Service de Rheumatologie, C.H.U. la Cavale Blanche, Brest, France; [2]Service de Rheumatologie, C.H.U. Hôpital Roger Salengro, Lille, France; [3]Cemka Eval, Bourg-la-Reine, France; [4]PHISQUARE, Besançon, France; [5]Service d'Immuno-Rheumatologie, C.H.U. Lapeyronie, Montpellier, France; [6]Mapi, Lyon, France; [7]UCB Pharma, Colombes, France; [8]UCB Pharma, Monheim, Germany

SAT0050

OBJECTIVE

- To evaluate the early performance of composite scores and HAQ-DI for predicting treatment outcome at 1 year in patients with rheumatoid arthritis in everyday clinical practice in France.

BACKGROUND

- There are currently no biomarkers capable of predicting treatment response and long-term benefit in rheumatoid arthritis (RA).
- Current practice guidelines emphasize the need to adapt treatment as a function of response: the treat-to-target strategy.[1]
 - In this context, composite scores and patient outcome measures are useful to identify treatment response/failure.
 - In particular, early prediction of treatment failure is fundamental to avoid unnecessary exposure to ineffective medication.
 - Identifying early treatment failure is potentially more useful than identifying persistent treatment response, since it enables treatment to be switched in a timely fashion, as recommended for a treat-to-target strategy.
- Several post-hoc analyses of clinical trials show that treatment failure at 3 months (M3) predicts treatment failure at 12 months (M12).[1,2]
- However, to our knowledge, there is no information from prospective studies on how markers of treatment failure would perform in everyday clinical practice.

METHODS

Design

- This is a 3-year longitudinal, prospective, observational, multicenter study of patients with RA starting certolizumab pegol (CZP) alongside standard therapy in France.
- Physicians were recruited from an exhaustive list of hospital rheumatologists and internal medicine specialists in France.
- Following agreement to participate in the study, each physician was expected to enroll the next three consecutive patients attending a consultation who fulfilled the eligibility criteria.
- Patients were expected to receive CZP treatment according to the Summary of Product Characteristics.
- Patients were evaluated at baseline (Week 0) and at 3-monthly routine consultations thereafter.
- Disease activity was assessed at each visit using the Clinical Disease Activity Index (CDAI);[3,4] Disease Activity Score-28 joint based on erythrocyte sedimentation rate (DAS28 [ESR]),[5] and the Health Assessment Questionnaire-Disability Index (HAQ-DI).[6]
- Data from a pre-specified interim analysis at M12 were used.

Patients

- Adult patients with moderate to severe, active RA, who were starting first-line treatment with CZP and had provided informed consent, were included in this study.

Data analysis

- The predictability of early treatment failure measures at M3 with respect to late treatment failure at M12 was assessed. Early failure and late failures are defined in **Table 1** for CDAI, DAS28(ESR) and HAQ-DI.
- For each evaluation, sensitivity, specificity, positive predictive value (PPV) and negative predictive value (NPV) were determined with 95% confidence intervals (CIs) **(Table 1)**.
- The PPV, assessing the probability that treatment failure at M12 is observed among all early failures at M3, was considered the principal analysis criterion.

Figure 1. Patient disposition

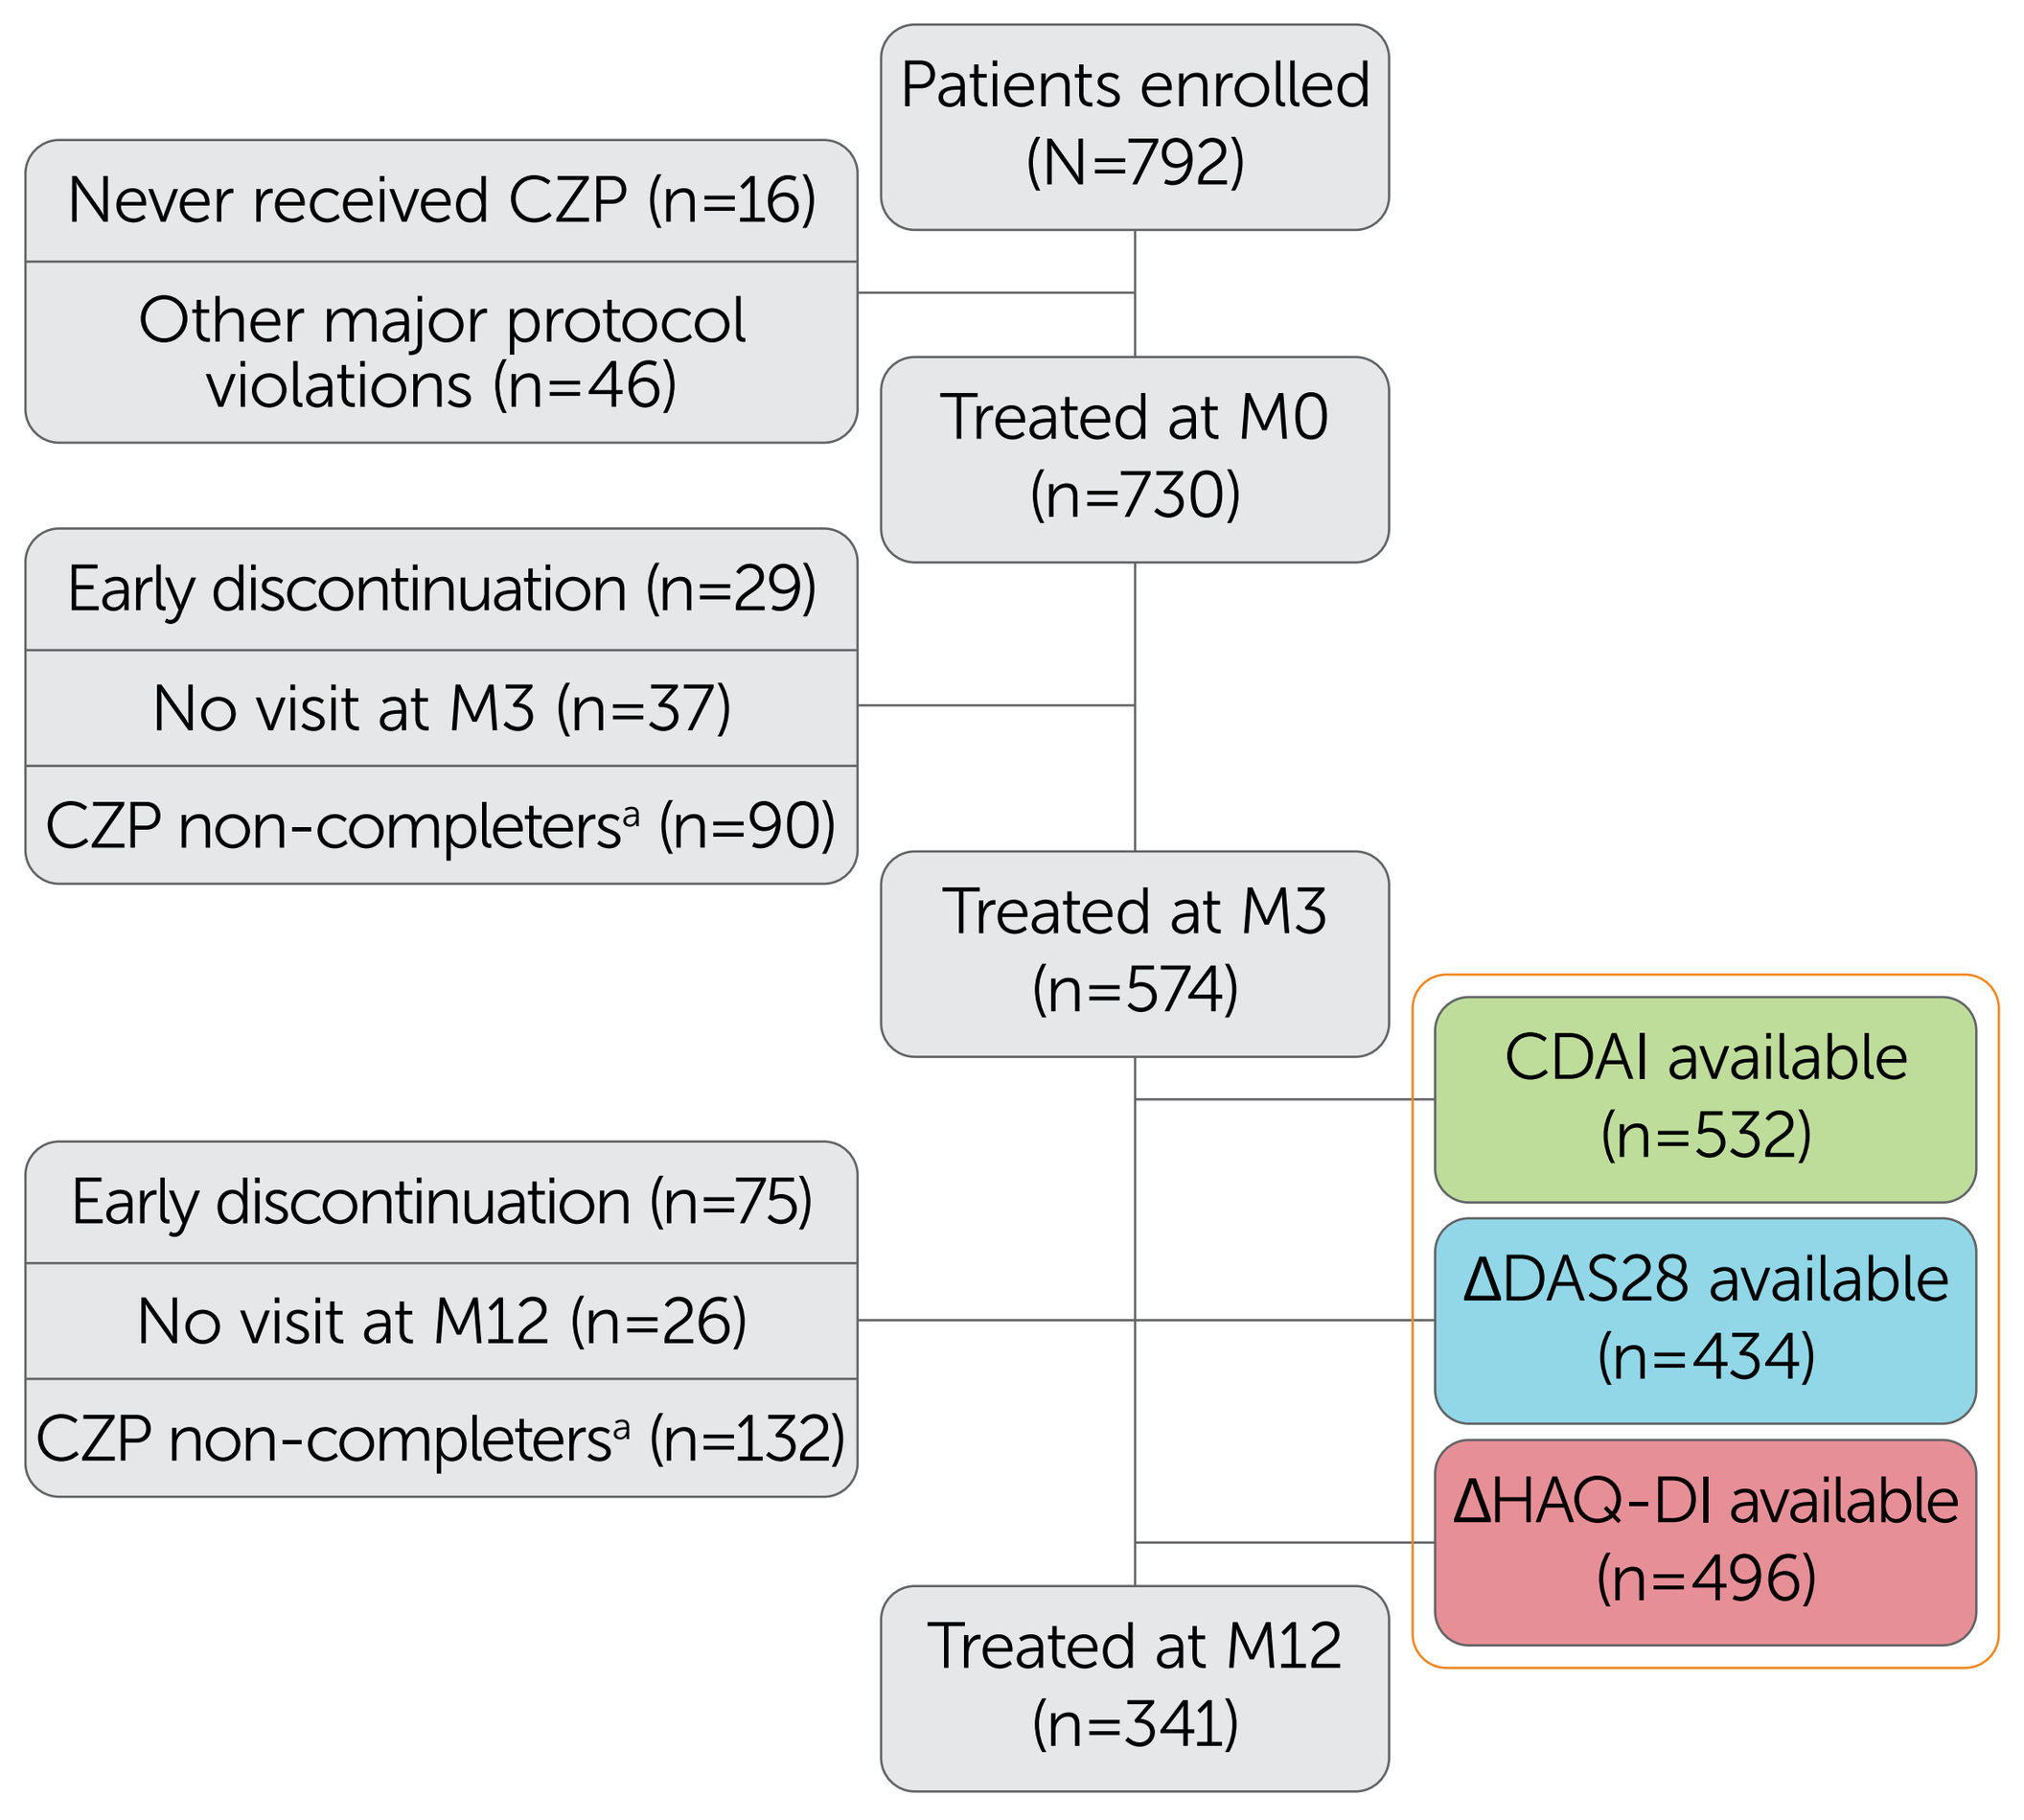
Patients enrolled (N=792)
Never received CZP (n=16)
Other major protocol violations (n=46)
Treated at M0 (n=730)
Early discontinuation (n=29)
No visit at M3 (n=37)
CZP non-completers[a] (n=90)
Treated at M3 (n=574)
CDAI available (n=532)
ΔDAS28 available (n=434)
ΔHAQ-DI available (n=496)
Early discontinuation (n=75)
No visit at M12 (n=26)
CZP non-completers[a] (n=132)
Treated at M12 (n=341)

[a]Includes patients who permanently discontinued CZP treatment before visit, or who temporarily discontinued CZP treatment for more than 2 weeks before the M3 visit, or more than 8 weeks before the M12 visit.

Table 1. Definitions used in the study

Outcome at M3	CDAI	DAS28(ESR)	HAQ-DI
Early treatment failure	Score >22[3]	Decrease of ≤1.2[3,7]	Decrease of <0.22[8]
Outcome at M12	**CDAI**	**DAS28(ESR)**	**HAQ-DI**
Late treatment failure	Score >10[9]	Score >3.2[3]	Score >0.5

Other definitions used in this study: **Sensitivity**: Among all patients in late treatment failure at M12, the probability that early non-response is observed at M3; **Specificity**: Among all patients with late treatment response at M12, the probability that early treatment response is observed at M3; **Positive predictive value (PPV)**: Among all patients with early non-response at M3, the probability that late treatment failure is observed at M12; **Negative predictive value (NPV)**: Among all patients with early response at M3, the probability that late treatment response is observed at M12.

Table 2. Baseline characteristics

	N=730
Age at inclusion (years)	
Mean ±SD	55.0 ±13.1
Female, n (%)	569 (77.9%)
Disease duration (years)	(n=728)
Mean ±SD	8.79 ±8.79
Tender/painful joint count (28 joints)	(n=721)
Mean ±SD	8.93 ±7.04
Swollen joint count (28 joints)	(n=719)
Mean ±SD	5.64 ±5.05
CDAI score	(n=698)
Mean ±SD	25.72 ±12.35
DAS28(ESR) score	(n=643)
Mean ±SD	4.84 ±1.27
HAQ-DI total score	(n=692)
Mean ±SD	1.27 ±0.69
Rheumatoid Factor	(n=378)
Positive (n, %)	263 (69.6%)
Anti-CCP antibodies	(n=396)
Positive (n, %)	292 (73.7%)
Prior treatments	(n=730)
No DMARDs	8 (1.1%)
Synthetic DMARDs	712 (97.5%)
Methotrexate	682 (93.4%)
Biological DMARDs	240 (32.9%)
Anti-TNF agents	221 (30.3%)
NSAIDs	403 (55.2%)
Glucocorticoids	562 (77.0%)
Concomitant anti-rheumatic drug treatments[a]	(n=730)
Synthetic DMARDs	471 (64.5%)
Methotrexate	391 (53.6%)
Leflunomide	63 (8.6%)
Hydroxychloroquine	21 (2.9%)
Sulfasalazine	17 (2.3%)
NSAIDs	258 (35.3%)
Glucocorticoids	373 (51.1%)

[a]At CZP initiation. CCP: cyclic citrullinated peptide; DMARD: disease-modifying antirheumatic drug; NSAID: non-steroidal anti-inflammatory drug; SD: standard deviation; TNF: tumour necrosis factor.

- Patients with no data available at M3 (missing visit or missing data), or no longer taking CZP at M3, were excluded from these analyses.
- Missing data at M12 were imputed by linear interpolation or last observation carried forward (LOCF). Failure was imputed at M12 in cases of permanent CZP discontinuation between M3 and M12.

RESULTS

Participating physicians

- A total of 3677 physicians were contacted in June 2011, of whom 365 (9.9%) agreed to participate in the study, and 176 (4.8%) recruited ≥1 patient: 170 rheumatologists and 6 internal medicine specialists.

Table 3. Early (M3) and late (M12) treatment failure rates

CDAI at M3	CDAI at M12: >10 (Late treatment failure)	≤10 (No late treatment failure)	Total
>22 (Early treatment failure)	79	10	89 (16.7%)
≤22 (No early treatment failure)	205	238	443 (83.3%)
Total	284 (53.4%)	248 (46.6%)	532
	Sensitivity=27.8%, 95% CI: 22.8–33.2 Specificity=96.0%, 95% CI: 93.0–97.9 PPV=88.8%, 95% CI: 81.0–94.1 NPV=53.7%, 95% CI: 49.1–58.3		

ΔDAS28 at M3	DAS28 at M12: >3.2 (Late treatment failure)	≤3.2 (No late treatment failure)	Total
≤1.2 (Early treatment failure)	136	61	197 (45.4%)
>1.2 (No early treatment failure)	102	135	237 (54.6%)
Total	238 (54.8%)	196 (45.2%)	434
	Sensitivity=57.1%, 95% CI: 50.8–63.3 Specificity=68.9%, 95% CI: 62.2–75.0 PPV=69.0%, 95% CI: 62.3–75.2 NPV=57.0%, 95% CI: 50.6–63.1		

ΔHAQ-DI at M3	HAQ-DI at M12: >0.5 (Late treatment failure)	≤0.5 (No late treatment failure)	Total
<0.22 (Early treatment failure)	175	57	232 (46.8%)
≥0.22 (No early treatment failure)	154	110	264 (53.2%)
Total	329 (66.3%)	167 (33.7%)	496
	Sensitivity=53.2%, 95% CI: 47.8–58.5 Specificity=65.9%, 95% CI: 58.5–72.7 PPV=75.4%, 95% CI: 69.6–80.6 NPV=41.7%, 95% CI: 69.6–80.6		

Sensitivity: Among all patients in late treatment failure at M12, the probability that early non-response is observed at M3; **Specificity**: Among all patients with late treatment response at M12, the probability that early treatment response is observed at M3; **Positive predictive value (PPV)**: Among all patients with early non-response at M3, the probability that late treatment failure is observed at M12; **Negative predictive value (NPV)**: Among all patients with early response at M3, the probability that late treatment response is observed at M12. Data are presented as number of patients in each case. 95% CIs calculated using the Jeffrey approximation.

Patients

- Overall, 792 patients were enrolled in the study, of whom 730 (92.2%) were analyzed.
- The number of patients available for predictability analyses ranged from 434 to 532 depending on CZP treatment status at M3, and on the availability of CDAI, DAS28 and HAQ-DI **(Figure 1)**.
- Baseline characteristics are reported in **Table 2**.

Predictivity

- Early and late treatment failure rates at M3 and M12, together with the indices of the predictive value of early treatment failure for predicting late treatment failure, are presented in **Table 3**.
- The highest PPV was observed for the CDAI compared to the DAS28(ESR) and the HAQ-DI **(Table 3)**.
- The PPV for CDAI was 88.8% (95% CI: 81.0–94.1), indicating that more than 80% of patients identified as non-responders at M3 failed to respond at M12.
- Specificity was also high (96.0%, 95% CI: 93.0–97.9), indicating that <5% of patients who achieved CDAI response at M12 were non-responders at M3.
- Of the 532/434/496 patients with CDAI/ΔDAS28/ΔHAQ-DI available, respectively, only 49 (9%)/38 (9%)/28 (6%) patients who would have been considered responders using alternative imputation were imputed as treatment failures by this analysis. Of these patients, 19 (46%)/15 (48%)/13 (59%) discontinued due to partial/lack of response and 15 (37%)/12 (39%)/7 (32%) due to intolerance/adverse events.

CONCLUSIONS

- This study was the first conducted under real-life conditions in France demonstrating a strong relationship between early and late treatment failure.
- The performance of CDAI as a predictor of long-term treatment failure was high, with more than 80% of CZP-treated patients experiencing treatment failure at 3 months also experiencing failure at 12 months.
- A stringent composite score like CDAI or DAS28(ESR) may be a reliable tool for predicting long-term treatment failure as early as 3 months, without the need for complementary biological tests, in a real-world cohort of patients affected by severe RA.

References

1. van der Heijde D. *et al.* J Rheumatol 2012;39(7):1326–33; **2.** Berenbaum F. *et al.* Joint Bone Spine 2017;doi:10.1016/j.jbspin.2017.01.011; **3.** Aletaha D. *et al.* Arthritis Res Ther 2005;7(4):R796–806; **4.** Aletaha D. and Smolen J. Clin Exp Rheumatol 2005;23(5 Suppl 39):S100–8; **5.** Prevoo M.L. *et al.* Arthritis Rheum 1995;38(1):44–8; **6.** Fries J.F. *et al.* Arthritis Rheum 1980;23(2):137–45; **7.** van Gestel A.M. *et al.* Arthritis Rheum 1996;39(1):34–40; **8.** Wells G.A. *et al.* J Rheumatol 1993;20(3):557–60; **9.** Zhang J. *et al.* Arthritis Care Res 2011;63(12):1672–9.

Author Contributions

Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: AS, RMF, FF, JM, BC, PH-M, JMJ, JD, GC; Drafting of the publication, or revising it critically for important intellectual content: AS, RMF, FF, JM, BC, PH-M, JMJ, JD, GC; Final approval of the publication: AS, RMF, FF, JM, BC, PH-M, JMJ, JD, GC.

Author Disclosures

AS: Consultant for: UCB Pharma; RMF: Consultant for: UCB Pharma; FF: Consultant for: UCB Pharma; JM: None declared; BC: Grant/research support from: Merck Pfizer Inc, Roche-Chugai, Consultant for: Merck, Pfizer, Roche-Chugai, UCB Pharma, Bristol-Myers Squibb, Celgene, Eli Lilly, Speaker's bureau: Merck, Pfizer, Roche-Chugai, UCB Pharma, Bristol-Myers Squibb, Celgene, Eli Lilly, Novartis; PH-M: None declared; J-MJ: Employee of: UCB Pharma; JD: Employee of: UCB Pharma; GC: Employee of: UCB Pharma.

Acknowledgements

This study was funded by UCB Pharma. We thank the patients and their caregivers in addition to the investigators and their teams who contributed to this study. Editorial services for this poster were provided by Costello Medical Consulting. All costs associated with development of this poster were funded by UCB Pharma.

Presented at EULAR 2017 | Madrid, Spain | 14–17 June 2017 | Previously presented at SFR 2016: Porte de Versailles, France | 11–13 December 2016